Genetically modified chicory brings hope to African malaria patients

Harro Bouwmeester[1] and Herwig Jansen[2]

A new solution for malaria?

Dafra Pharma International NV and Plant Research International (PRI) have started new research to produce artemisinin via genetically modified chicory plants. This research should result in inexpensive, large-scale production of artemisinin. Artemisinin is a basic raw material used in ACTs (Artemisinin based Combination Therapies), the latest generation and most effective antimalaria treatment according to the WHO (World Health Organization of the UN).

Malaria and ACTs

According to the WHO some 300 to 500 million malaria cases per year are reported worldwide. Each year this results in the death of 1.5 to 2 million people, of which 90% occur in Africa [1]. Rapid treatment with an ACT could save many lives. Artemisinin, however, is relatively expensive as its only source is a plant, *Artemisia annua*, growing mainly in China and Vietnam and only producing low yields. Therefore, an ACT these days easily costs ten times more than a treatment with e.g. chloroquine. This is too expensive for most African patients and this can only be solved if the price of the ACTs, and thus particularly the price of artemisinin, strongly decreases.

Biotechnological production of artemisinin

In earlier research in Wageningen the complete biosynthetic pathway of artemisinin was resolved (Fig. 1) [2]. In addition, the Wageningen scientists, headed by Prof. Harro Bouwmeester and Dr. Maurice Franssen, could demonstrate that chicory enzymes – normally involved in the biosynthesis of the bitter sesquiterpene lactones in chicory - are capable of performing reactions required for the biosynthesis of artemisinin [3]. Via a diversion of the biosynthesis of bitter compounds Prof. Bouwmeester will produce the chemical precursor for artemisinin (dihydroartemisinic acid) in the roots of chicory. The group of Prof. Bouwmeester has shown in a wide range of plant species that diversion of the biosynthesis of terpenes can be carried out very efficiently [4].

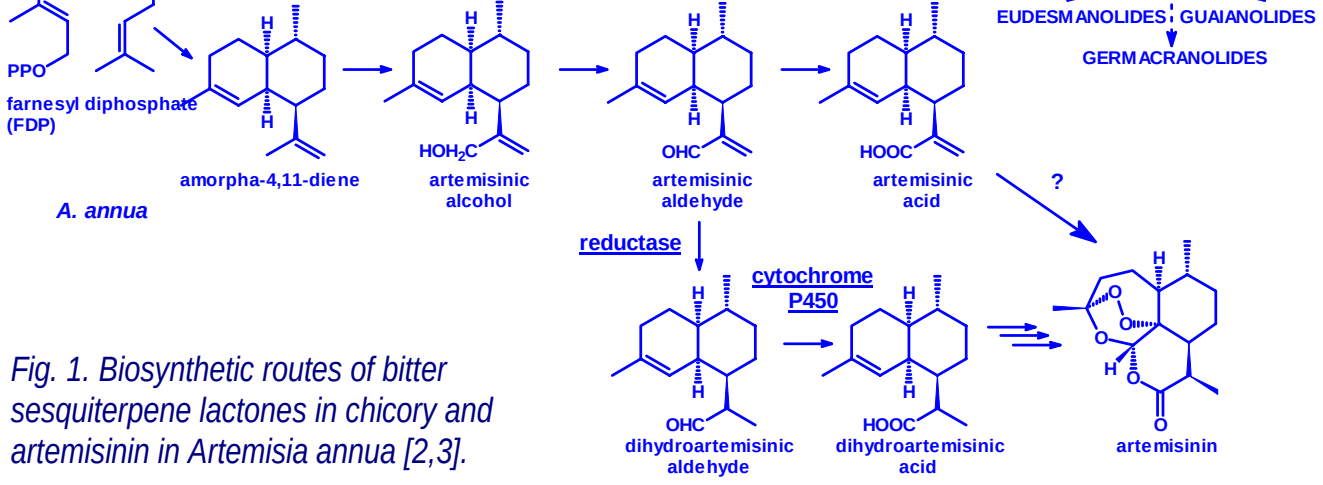
chicory
(+)-germacrene A
CH_2OH
CHO
COOH
(+)-costunolide
cytochrome P450
EUDESMANOLIDES
GUAIANOLIDES
GERMACRANOLIDES
PPO
farnesyl diphosphate (FDP)
amorpha-4,11-diene
HOH_2C
artemisinic alcohol
OHC
artemisinic aldehyde
HOOC
artemisinic acid
?
A. annua
reductase
cytochrome P450
dihydroartemisinic aldehyde
dihydroartemisinic acid
artemisinin

Fig. 1. Biosynthetic routes of bitter sesquiterpene lactones in chicory and artemisinin in Artemisia annua [2,3].

Why chicory?

We have chosen inulin chicory as artemisinin production platform because it is a well-established industrial crop for a.o. non-food applications. This means that the entire chain of large-scale agricultural production, including extraction, is already operational. Dafra Pharma International NV has the chemical expertise required for the conversion, after extraction, of the precursor into artemisinin that is directly suitable for the production of ACTs.

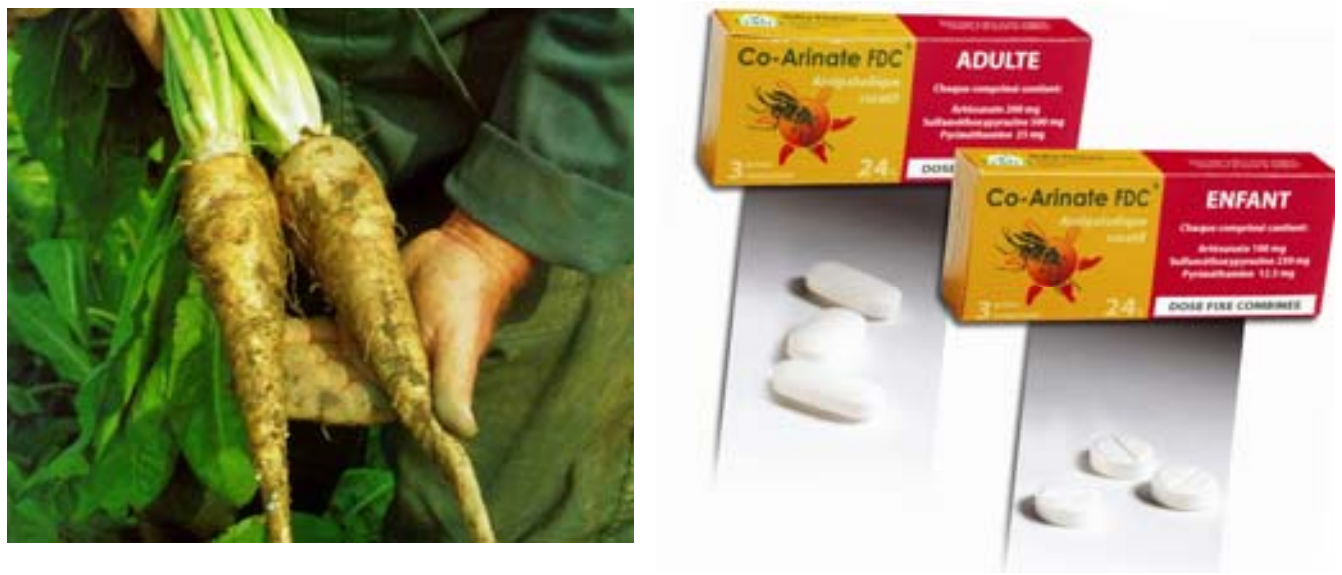
Co-Arinate FDC
ADULTE
Co-Arinate FDC
ENFANT
24
3

Fig. 2. Inuline chicory (left), a crop highly adapted to industrial production and processing, a new source for anti-malarial Artemisinin based Combination Therapies (ACTs).

Conclusion

Together, this research will result in the large-scale production of artemisinin which should lead to cheap, but high-quality, effective and safe antimalaria treatments (ACTs). It is anticipated that this new inexpensive raw material will enable Dafra Pharma International NV to place its ACTs on the market for half a dollar per adult treatment.

References

1. WHO, 2005, World malaria report
2. Bertea et al., 2005. Planta Medica 71: 40-47
3. de Kraker et al., 2003. Tetrahedron 59: 409-418
4. Kappers et al., 2005. Science 309: 2070-2072
5. Gibson et al., 1994

Fig. 3. Schematic representation of inuline extraction facility. In such facilities the bitter compounds and other unwanted metabolites are efficiently removed. That part of the process will be used to extract the artemisinin precursor. From [4]

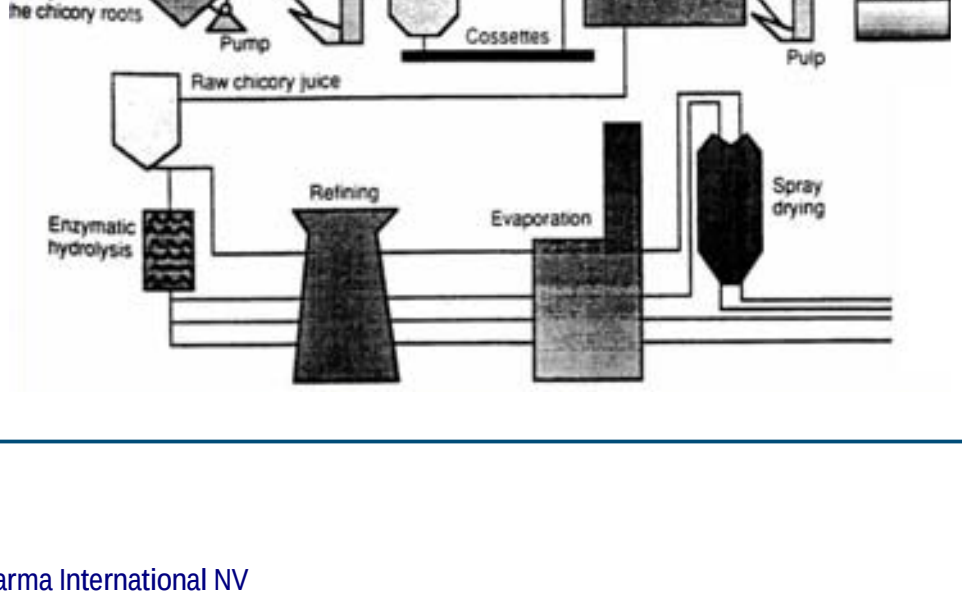
Washer
Slicer
Diffuser
Reception of the chicory roots
Pump
Cossettes
Dryer
Pulp
Raw chicory juice
Refining
Evaporation
Spray drying
Enzymatic hydrolysis

[1]Plant Research International
P.O. Box 16, 6700 AA Wageningen, The Netherlands
Tel: +31 317 475882 - Fax: +31 317. 41 80 94
E-mail: harro.bouwmeester@wur.nl

[2]Dafra Pharma International NV
Slachthuisstraat 30/7, 2300 Turnhout, Belgium
Tel: +32 (0)14 61 78 20 - Fax: +32 (0)14 47 29 90
E-mail: ellen.jansen@dafra.be